Mitigation of Injection Site Reactions after Subcutaneous Administration of Dalcinonacog Alfa (DalcA) in Hemophilia B Using Preclinical Models

Natacha Le Moan[1], Lauren Kelly[1], Eric Merle[2], Pascal Descargues[2], Nicolas Gaudenzio[2], Hugo Gagnon[3], Anju Chatterji[1], and Grant E. Blouse[1]

[1]Catalyst Biosciences, South San Francisco, CA, USA [2]Genoskin, Inc. 4 Technology way, Salem, MA, 01970, USA [3]Phenoswitch Biosciences, 975 Rue Léon-Trépanier, Sherbrooke, QC J1G 5J6, Canada

CATALYST BIOSCIENCES

Abstract Number: PB0453

Introduction

Prophylactic subcutaneous (SQ) administration of dalcinonacog alfa (DalcA), an engineered recombinant factor IX (FIX) variant with 22-fold enhanced potency compared with BeneFIX, provides stable and protective therapeutic FIX levels in individuals with Hemophilia B. Phase 1 and 2b studies demonstrated DalcA was well tolerated and efficacious, showing sustained FIX levels and no bleeding events from therapy start through washout. However, some subjects reported mild-to-moderate injection site reactions (ISR) consisting of pain and/or redness following initial SQ injections. To investigate the underlying ISR mechanism, we examined cutaneous microanatomic, cellular, and proteomic changes after SQ DalcA administration in the HypoSkin® injectable human skin biopsy platform (Genoskin, Salem, MA, USA) and Göttingen minipigs.

Reverse Translational Approach

Daily DalcA SQ injections for 8-14 days induced erythema and redness in minipigs comparable with the observed human ISR. Human skin biopsies treated with DalcA underwent proteomic analysis to assess if minipig ISR predictors could be identified in human skin 36 hours after SQ injection. ISR-like response classification in this *ex vivo* setting was based on observed histological modifications.

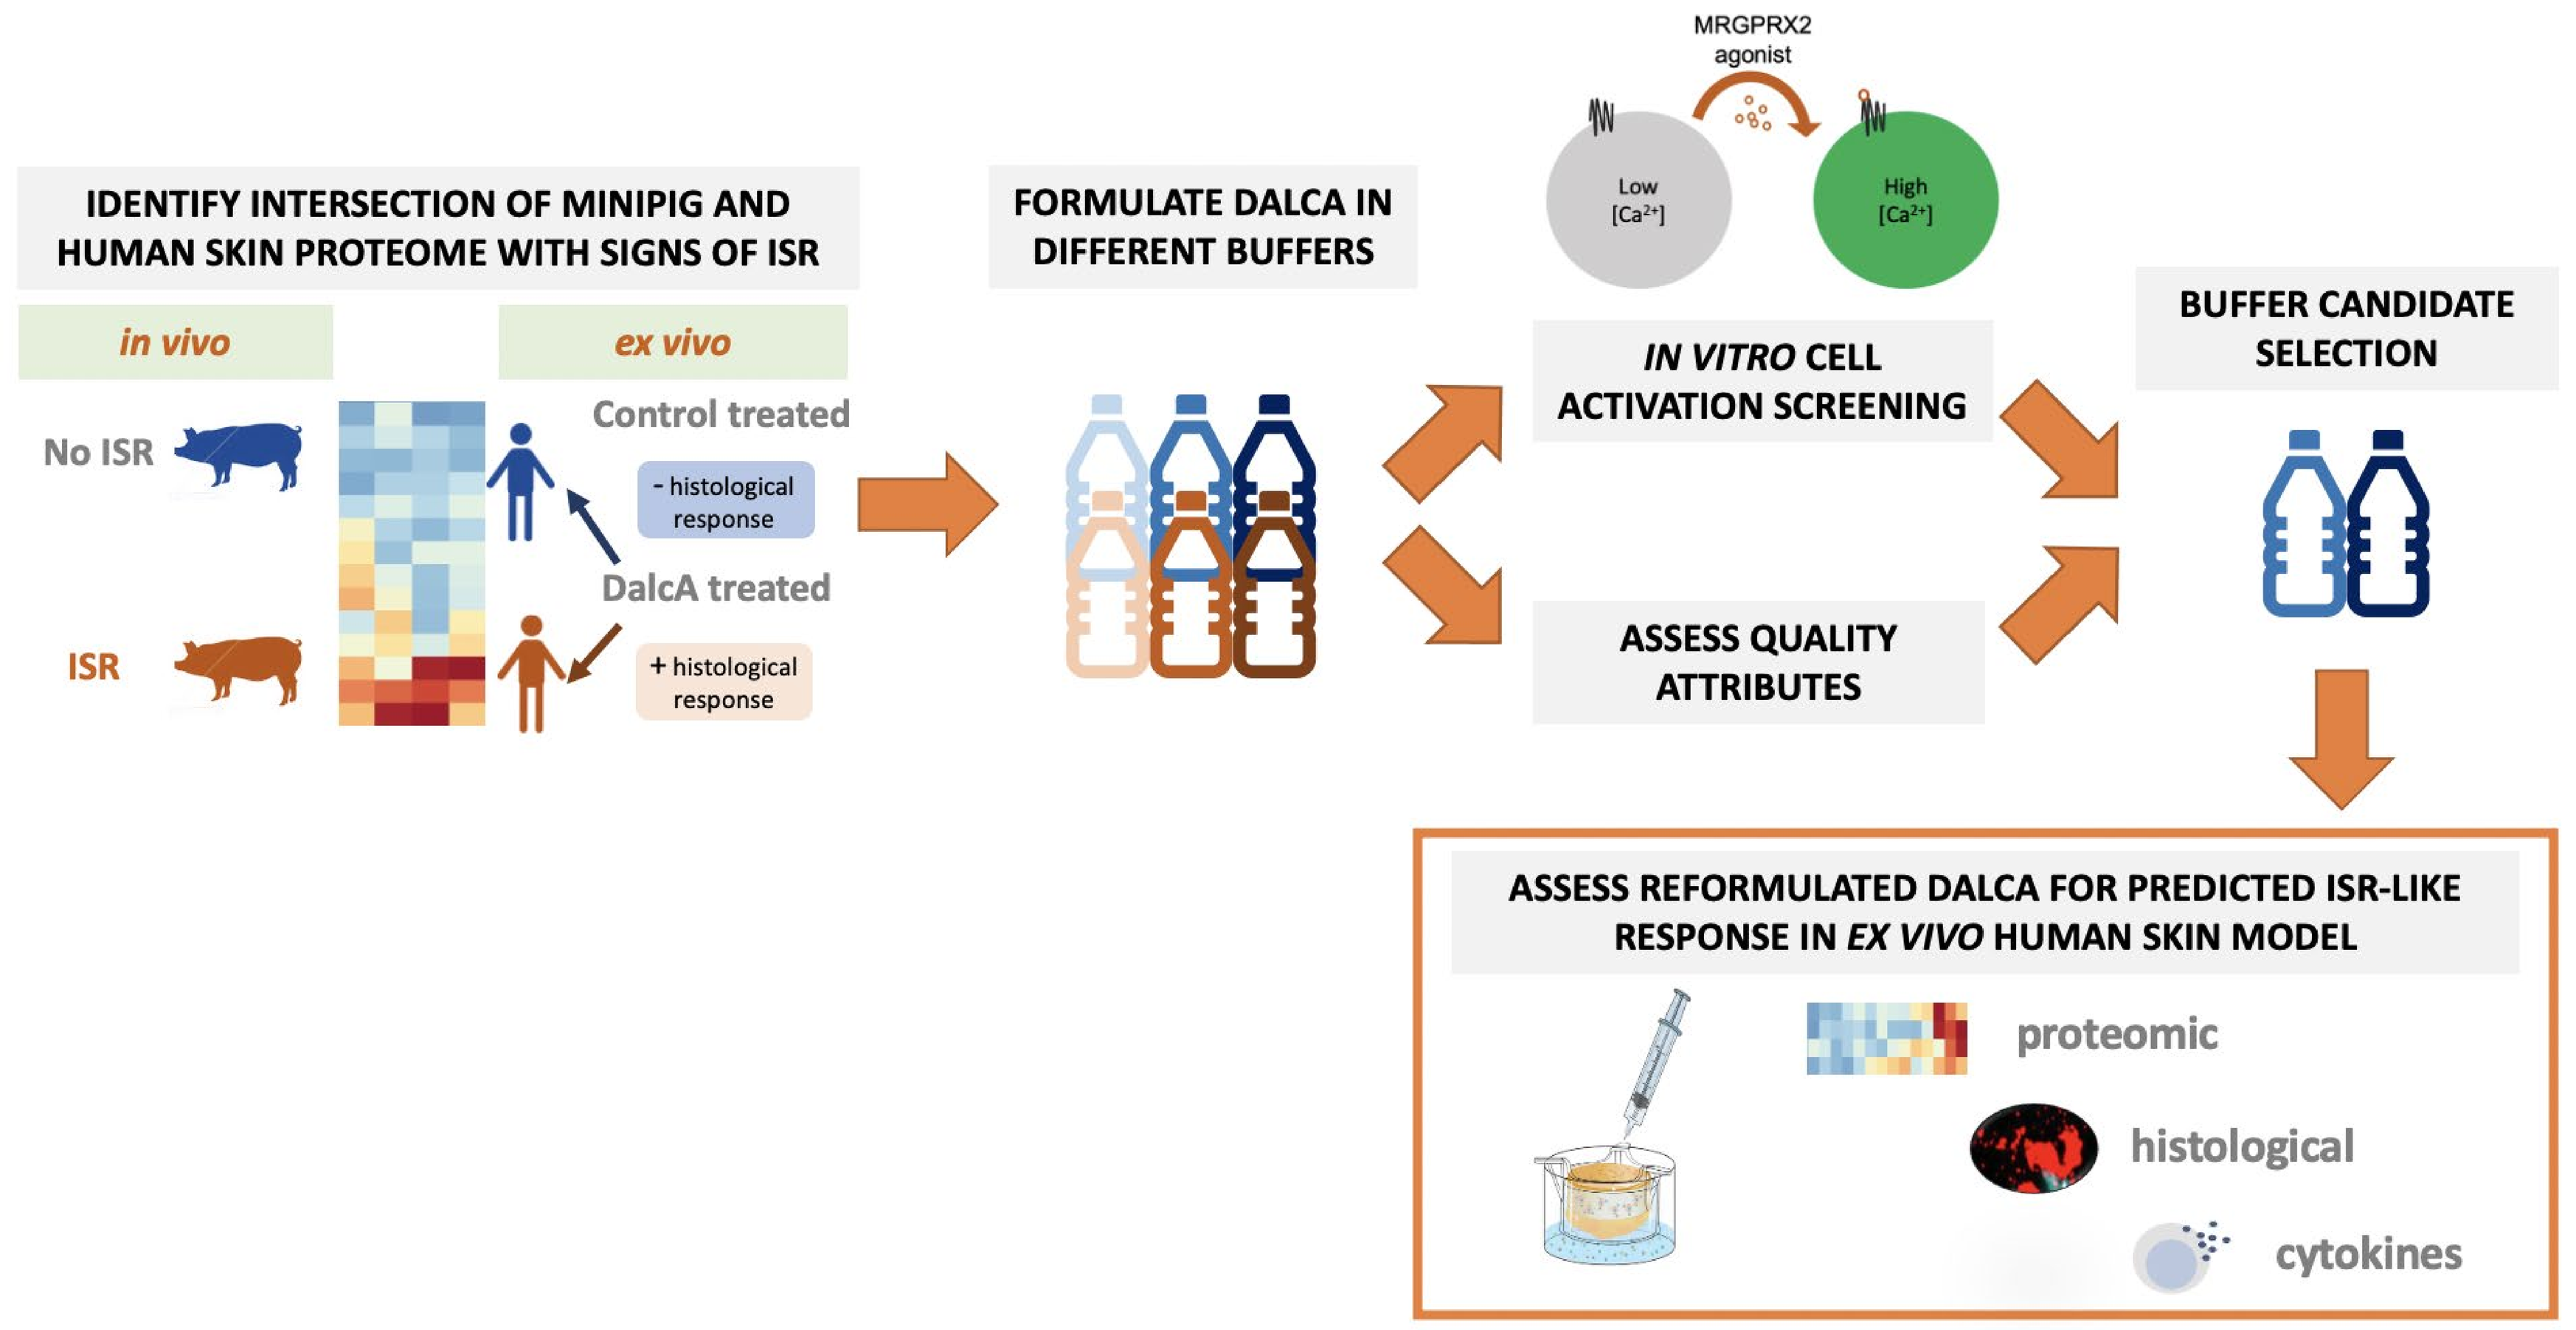
IDENTIFY INTERSECTION OF MINIPIG AND HUMAN SKIN PROTEOME WITH SIGNS OF ISR
in vivo
ex vivo
No ISR
ISR
Control treated
- histological response
DalcA treated
+ histological response
FORMULATE DALCA IN DIFFERENT BUFFERS
MRGPRX2 agonist
Low [Ca2+]
High [Ca2+]
IN VITRO CELL ACTIVATION SCREENING
ASSESS QUALITY ATTRIBUTES
BUFFER CANDIDATE SELECTION
ASSESS REFORMULATED DALCA FOR PREDICTED ISR-LIKE RESPONSE IN EX VIVO HUMAN SKIN MODEL
proteomic
histological
cytokines

Results

Table 1: ISR-like proteomic signature

UniProt ID	Gene	Up/Down regulated
O43707	ACTN4	down
P00738	HP	down
P01714	IGLV3-19	down
P35749-3	MYH11	down
P35749	MYH11	down
O14558	HSPB6	up
P00747	PLG	up
P04004	VTN	up
P07339	CTSD	up
P13796	LCP1	up
P29350	PTPN6	up

UniProt ID	Gene	Up/Down regulated
P29350	PTPN6	up
P38606	ATP6V1A	up
P52597	HNRNPF	up
P53999	SUB1	up
P78371	CCT2	up
Q15181	PPA1	up
Q15185	PTGES3	up
Q7Z7G0	ABI3BP	up
Q9H299	SH3BGRL3	up
Q9UFN0	NIPSNAP3A	up
Q9Y3Z3	SAMHD1	up

+From viewing the proteomic signatures in mini-pigs and human skin, 22 common proteomic regulation changes were associated with ISR-like responses from subcutaneously administered DalcA (Table 1)

+Boolean scoring based on this signature correctly identified 2/2 ISR-like and 5/6 non-ISR-like findings in human skin with a false-positive rate of ~16% and a false-negative rate of 0%, suggesting a strong biomarker profile for ISR

Ex vivo studies in the HypoSkin ® injectable human skin biopsy platform (Genoskin, Salem, MA, USA) showed that DalcA stimulated mast cell degranulation within 4 hours. *In vitro* studies further demonstrated that DalcA activates the Mas-related G protein-coupled receptor X2 (MRGPRX2) expressed by mast cells and associated with dermal ISR to a similar level as wild-type recombinant FIX. Since cationic drugs can stimulate MRGPRX2, we examined MRGPRX2 activation of DalcA when presented in alternative formulations.

Figure 1 : Methodology for MRGPRX2 activation screening

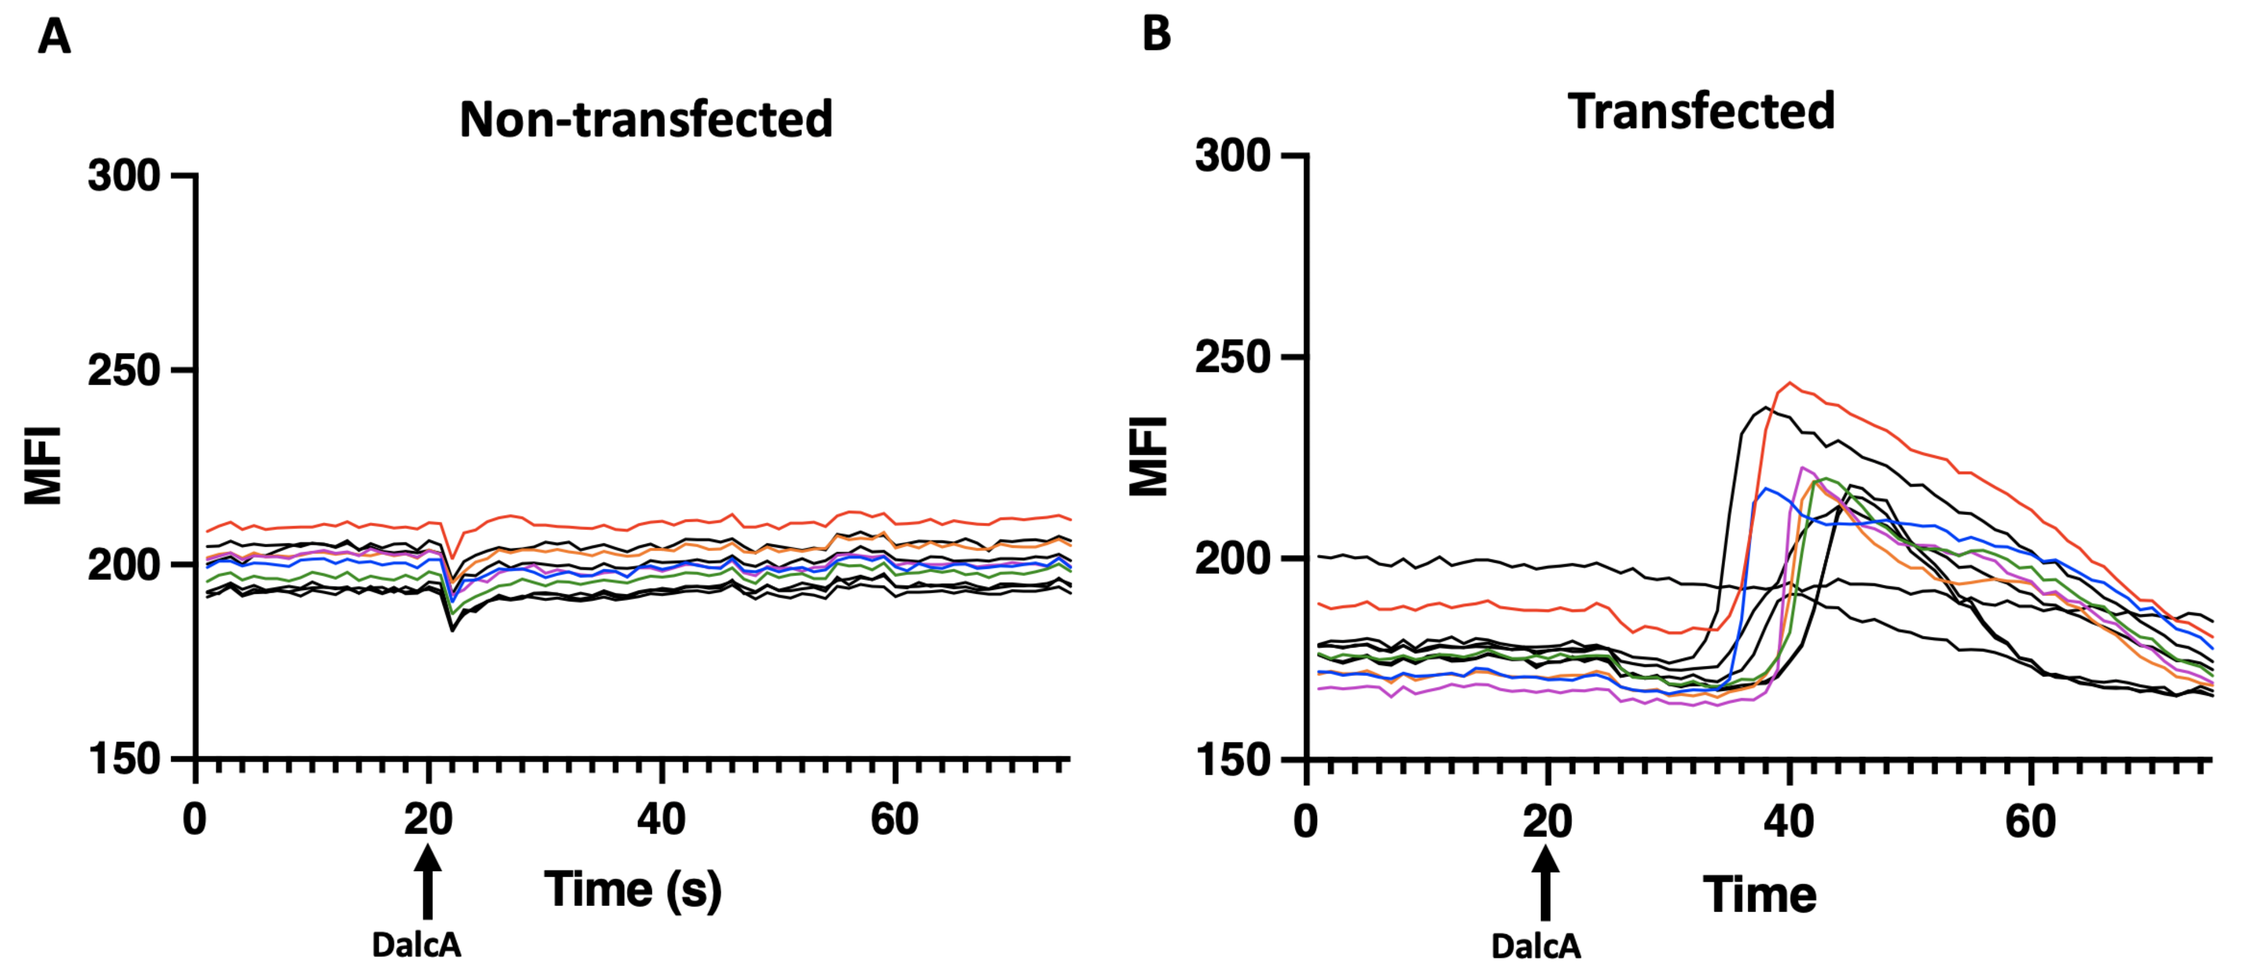
A
Non-transfected
MFI
300
250
200
150
0
20
40
60
Time (s)
DalcA
B
Transfected
MFI
Time
DalcA

Table 2: Buffer compositions

Formulation Code	PS80 (%)	pH
A1	0.004	6.5
A2	0.017	
A3	0.03	
B1*	0.004	6.8
B2	0.017	
B3	0.03	
C1	0.004	7.1
C2	0.017	
C3	0.03	

* B1 matches the target specifications of the classical DalcA formulation buffer

+HEK cells transfected to express the G-protein coupled receptor MRGPRX2 were exposed to DalcA drug substance in formulations with varying PS80 (Tween-80) and pH levels (Table 2)

+Example fluorescent signal traces for individual transfected (B) or non-transfected (A) cells demonstrate the fluorescent signal response of DalcA administration beginning at 20 seconds

Figure 2 : Buffer composition can influence MRGPRX2 activation *in vitro*

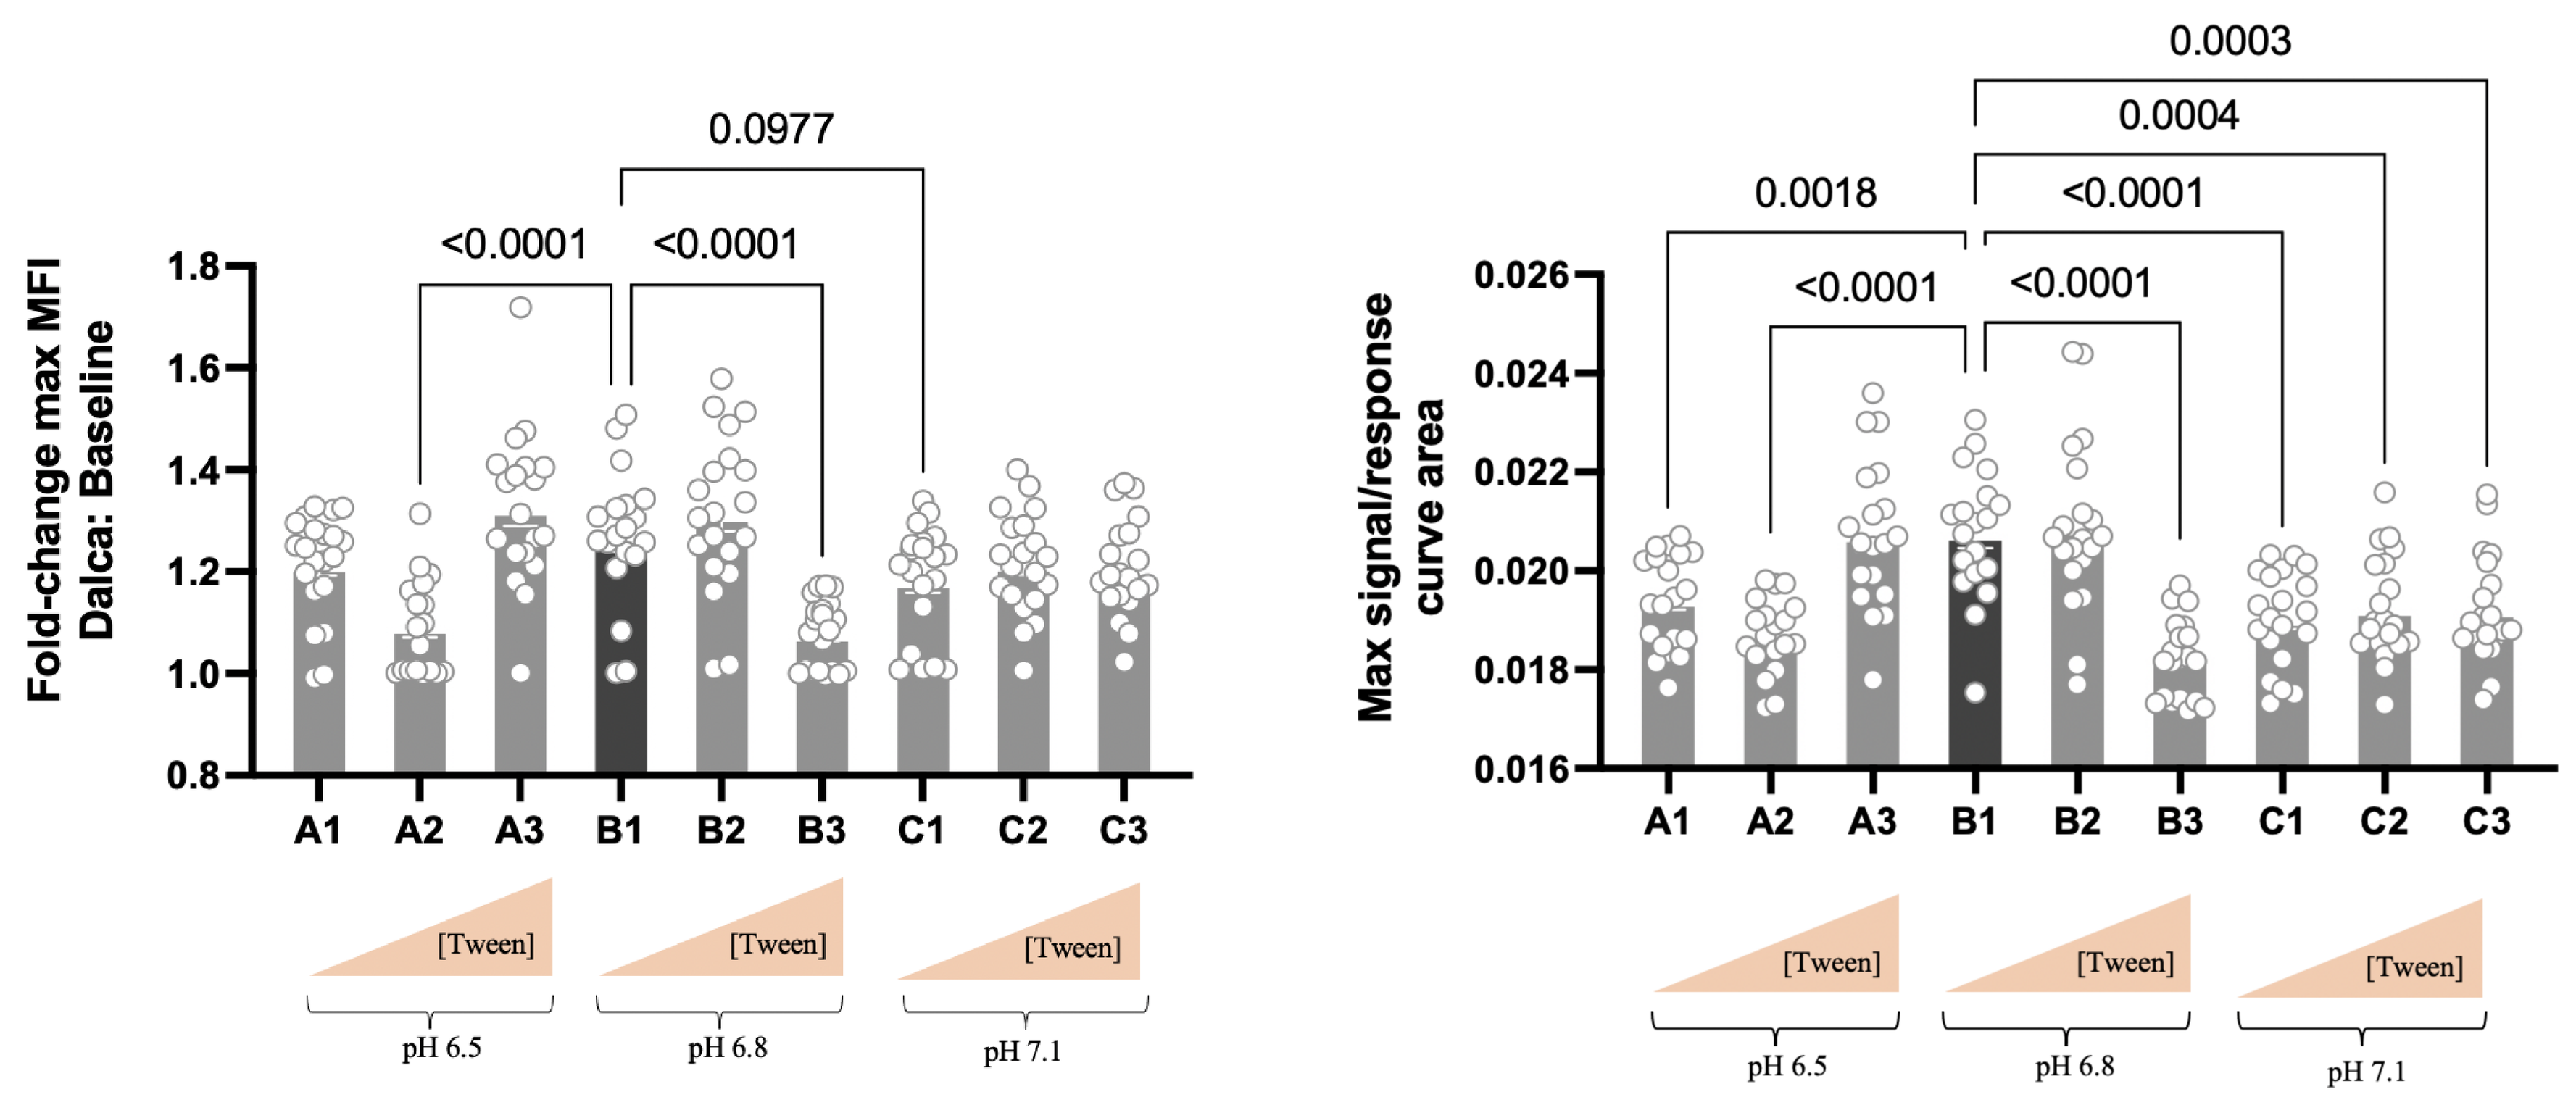
Fold-change max MFI Dalca: Baseline
0.0977
<0.0001
<0.0001
A1 A2 A3 B1 B2 B3 C1 C2 C3
[Tween]
pH 6.5
pH 6.8
pH 7.1
Max signal/response curve area
0.0003
0.0004
0.0018
<0.0001
<0.0001
<0.0001

+Peak signal intensity from MRGPRX2 activation was reduced when stimulated with DalcA in formulation buffers with high tween content (A2 & B3) or higher pH (C1) compared to target formulation (B1)

+Peak signal intensity corrected for total duration of response suggests a higher pH formulation can reduce MRGPRX2 activation *in vitro* at all tween contents (C1, C2, and C3) compared to B1

+All low pH or high tween-80 buffers (orange formulations in Table 2) demonstrated diminished stability attributes compared to the target formulation (B1) under accelerated storage conditions

Formulation buffers with higher pH and/or tween content reduced *in vitro* MRGPRX2-mediated cell activation, suggesting that optimizing the DalcA formulation buffer may reduce *in vivo* mast cell degranulation and downstream signaling of pain and dermal inflammation. We examined mast cell degranulation, cytokine secretion, and proteomic changes after SQ DalcA administration in Hyposkin human biopsies for a composite ISR risk profile.

Figure 3: Buffer composition can affect ISR-like response in human skin biopsies

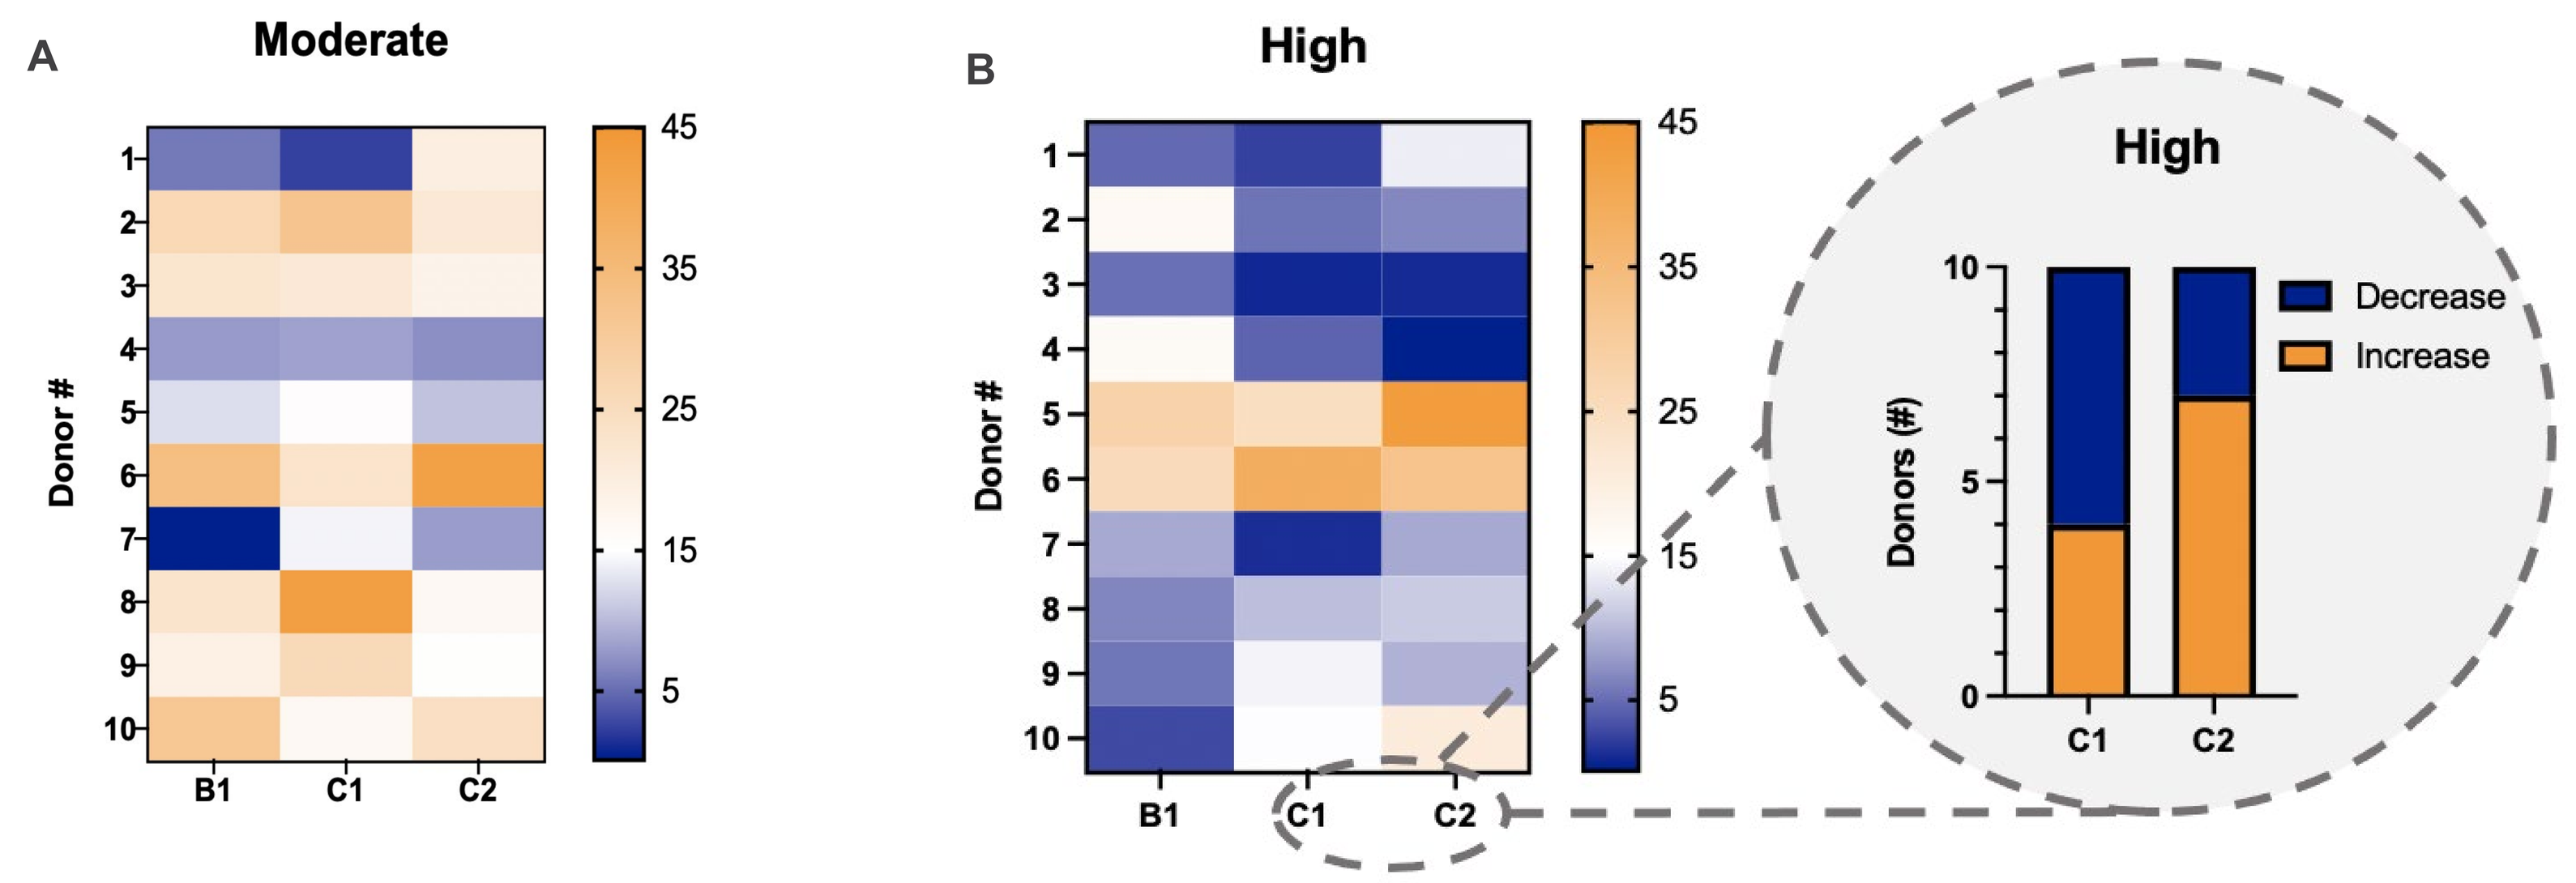
A
Moderate
Donor #
B1 C1 C2
B
High
High
Donors (#)
Decrease
Increase
C1 C2

+Percent mast cells observed with moderate (A) or high (B) degranulation 4 hours after SQ injection

+Relative to DalcA in B1, C1 stimulated fewer highly degranulating mast cells in most donors (insert)

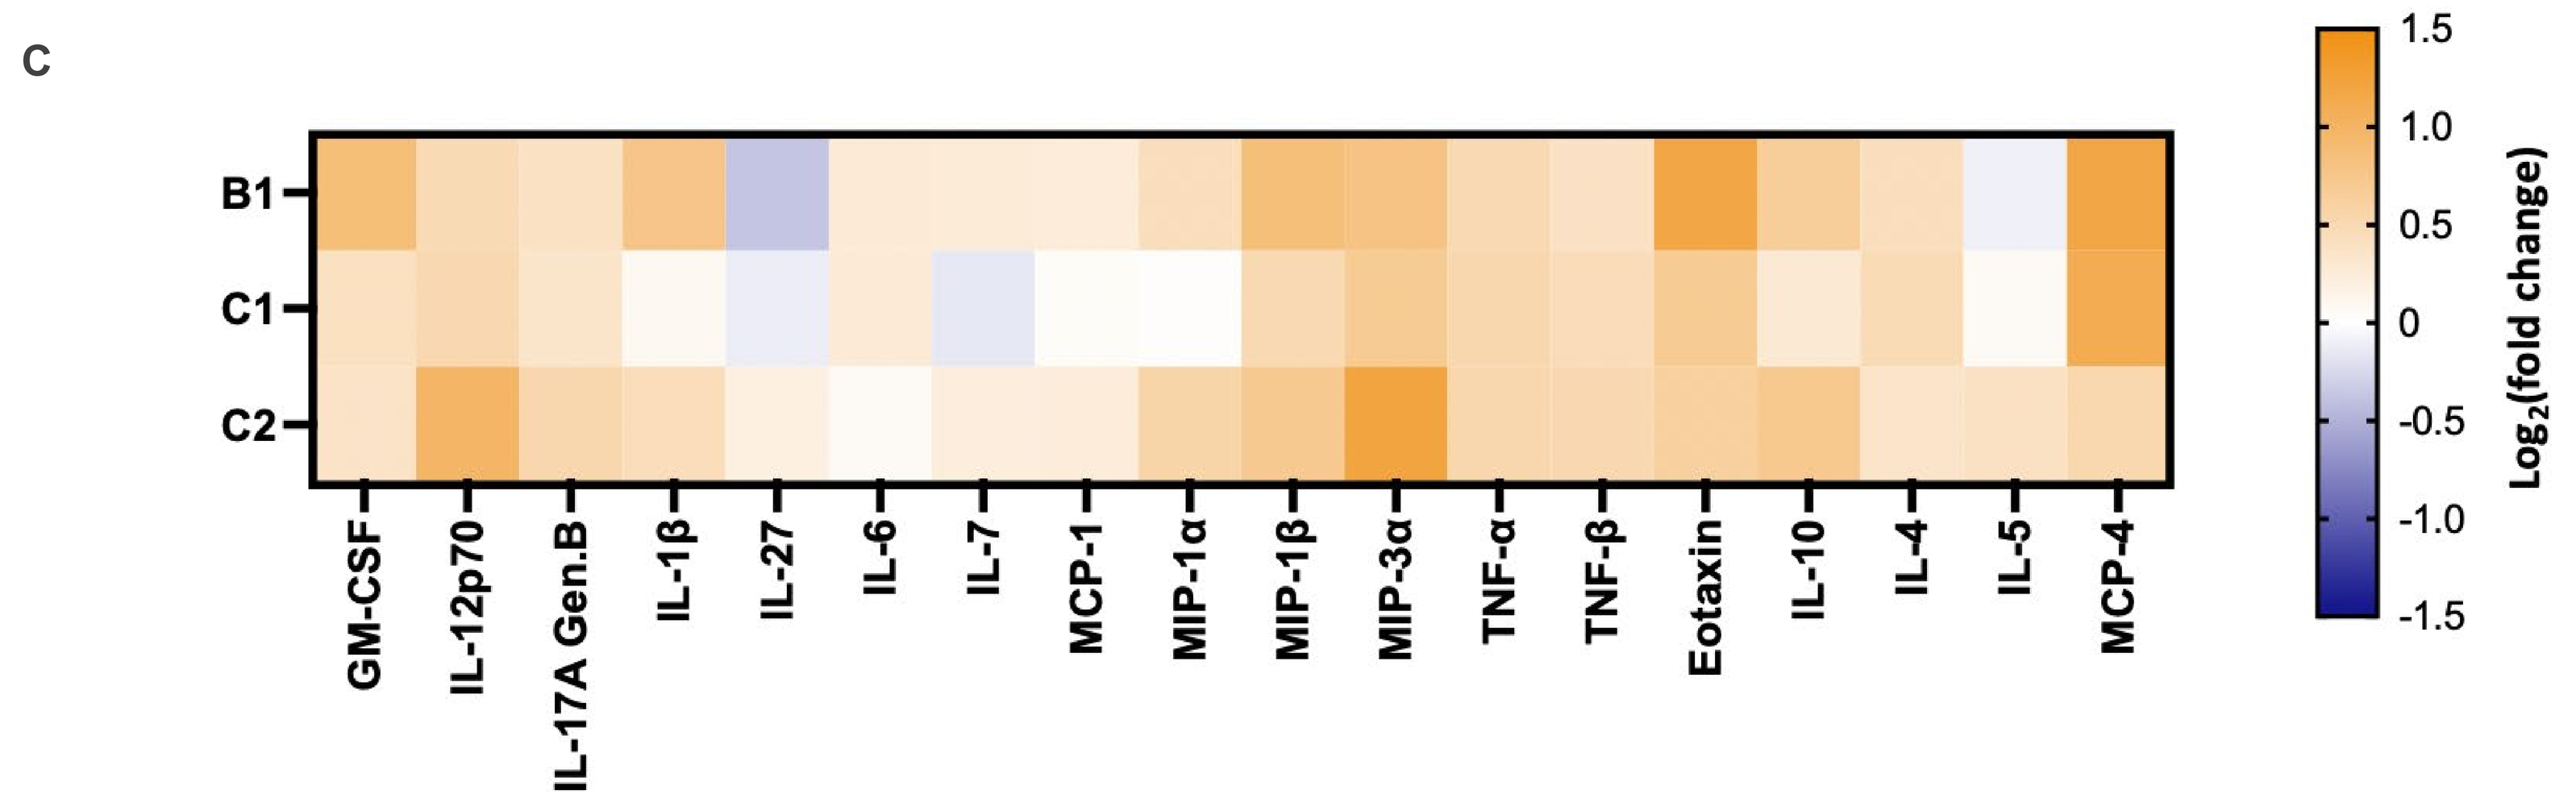
C
B1
C1
C2
GM-CSF
IL-12p70
IL-17A Gen.B
IL-1β
IL-27
IL-6
IL-7
MCP-1
MIP-1α
MIP-1β
MIP-3α
TNF-α
TNF-β
Eotaxin
IL-10
IL-4
IL-5
MCP-4
Log2(fold change)

+Treatment associated cytokine level was divided by the negative control for a relative fold change and averaged across donors (C)

+DalcA in buffer C1 performed similarly to the negative control for most cytokines for 8/10 donors

+DalcA formulated in buffer C2 stimulated a different cytokine response compared to both classical Dalca (B1) and the negative control in 4/8 donors

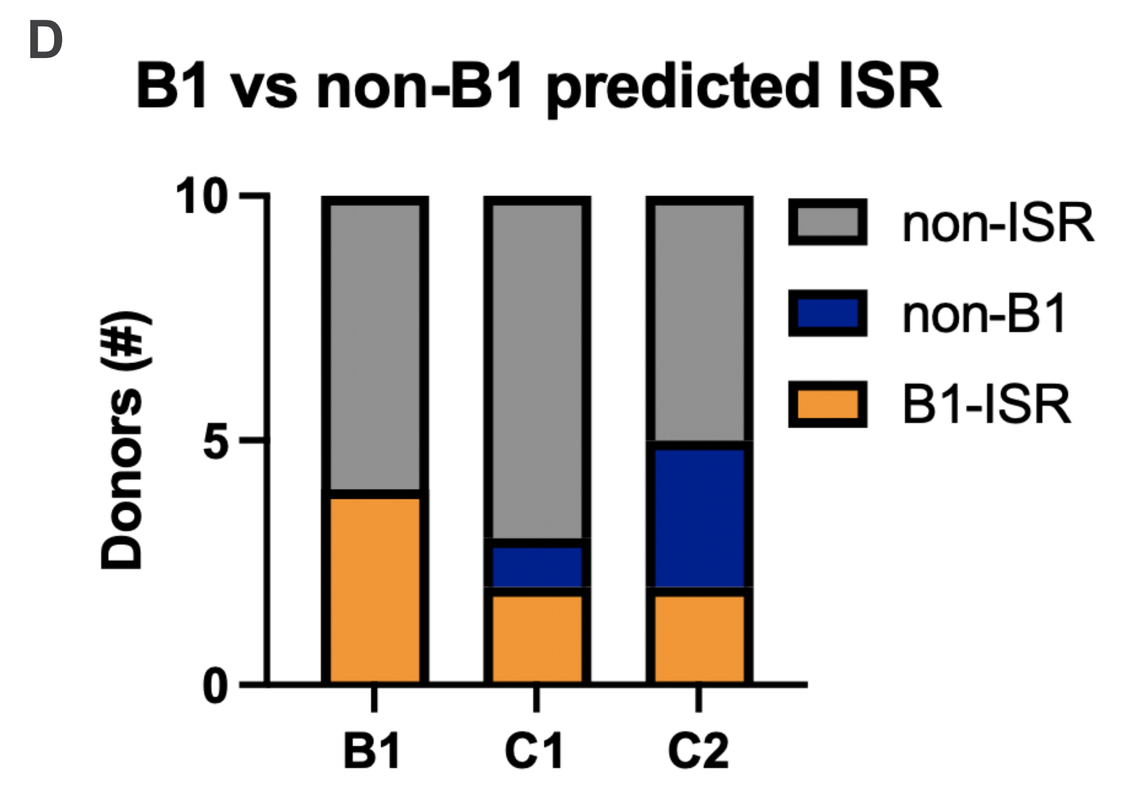
D
B1 vs non-B1 predicted ISR
Donors (#)
non-ISR
non-B1
B1-ISR
B1 C1 C2

+Consistent with clinical incidence findings, DalcA in B1 was predicted to elicit an ISR-like response based on proteomic signature changes in 4/10 donors (D)

+DalcA in C1 buffer reduced ISR-like findings to 3/10 donors

+DalcA formulated in C2 buffer increased the incidence to 5/10 donors, including 3 donors not predicted to experience ISR with DalcA in buffer B1 (“non-B1”)

Conclusions

+Formulation buffer composition affects DalcA MRGPRX2-mediated cell activation *in vitro*

+A biomarker signature predictive of minipig ISR was generated from the cross comparison of cutaneous proteomes of minipig and *ex vivo* human skin models

+Hyposkin platform enabled evaluation and selection of optimal formulation buffer expected to lower the risk of clinical ISR upon SQ DalcA injection