The Combination of Marzeptacog Alfa (Activated) or Eptacog Alfa (Activated) with Emicizumab Appears Comparable in the Thrombin Generation Test in Hemophilia A

Tom Knudsen, DVM, PhD[1], Brajesh Kumar, PhD[1], Frank Del Greco, MBA[1], Linda Neuman, MD[1], Howard Levy, PhD, MBBChir[1] and Grant E. Blouse, PhD[1]

[1]Catalyst Biosciences, South San Francisco, CA

Conclusion

- **MarzAA and NovoSeven exhibit comparable characteristics when spiked into HA plasma containing Hemlibra at clinically relevant concentrations and assessed by a standard thrombin generation assay**

Perspectives

- **Based on these data, MarzAA is expected to be safe in combination with Hemlibra. This provides a rationale for eventual clinical development of MarzAA as a SQ rescue therapy for Hemophilia A patients experiencing breakthrough bleeds while on Hemlibra prophylaxis**

Background

Hemophilia patients treated prophylactically with emicizumab (Hemlibra®) may experience break-through bleeds or require additional hemostatic coverage for procedures or surgery. Currently available therapies including rFVIIa (Eptacog Alfa (Activated); NovoSeven®) and aPCC (FEIBA®) have been used with Hemlibra to treat bleeding or when additional coverage is required. While NovoSeven appears safe in combination with Hemlibra (HAVEN 1 to 4 clinical trials), thrombotic events have been observed with concurrent use of FEIBA and Hemlibra. While safe and efficacious when used as directed, NovoSeven must be infused intravenously.

Ideally, adjunct therapy for patients on subcutaneous (SQ) prophylaxis with Hemlibra would also be SQ. Marzeptacog alfa (activated) (MarzAA) is a novel rFVIIa differentiated by increased potency and the ability to be administered SQ to achieve pharmacologically relevant plasma concentrations. MarzAA provides a potential solution to address this unmet need in hemophilia therapy.

+ MarzAA has two amino acid substitutions in the protease domain (Q286R and M298Q) that increase catalytic activity for FX activation in the presence and absence of tissue factor
+ Two additional substitutions in the EGF2 domain of the light chain (T128N and P129A) create an additional N-linked glycosylation site

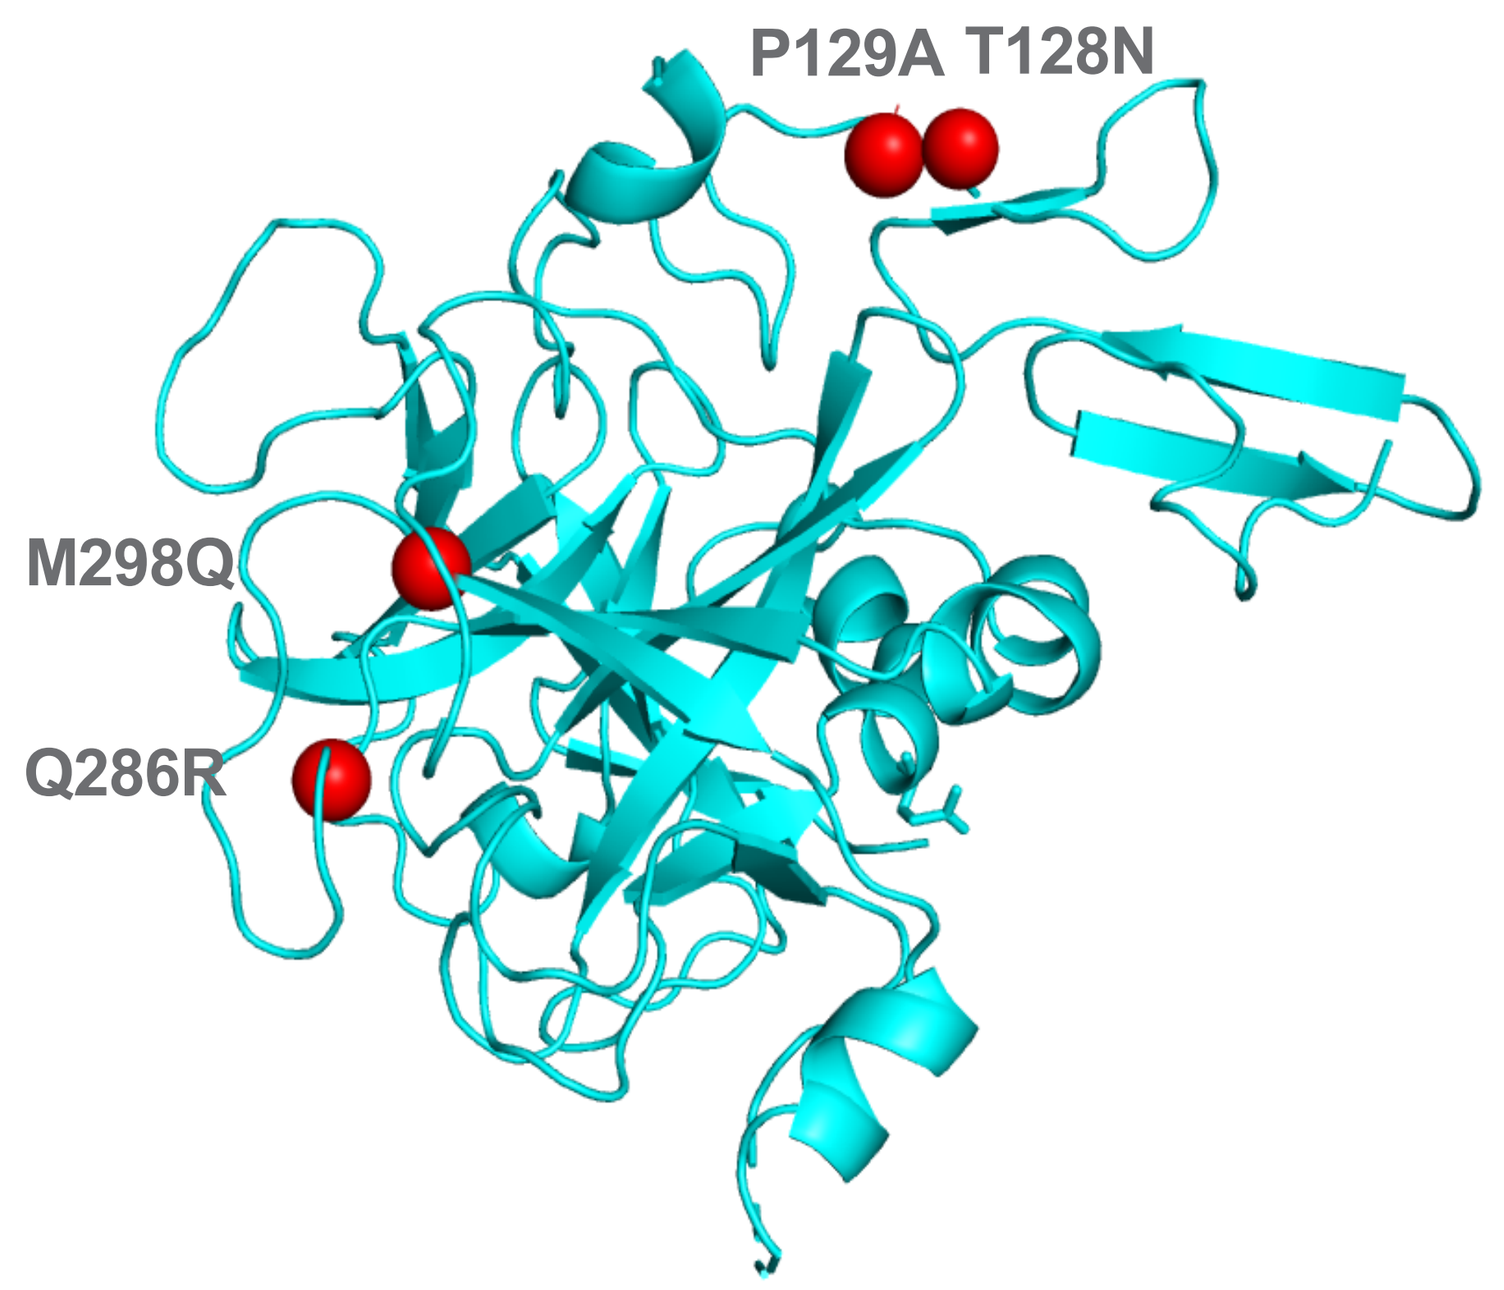
P129A T128N
M298Q
Q286R

Objective and Rationale

Primary objective

+ Evaluate the procoagulant potential of MarzAA, NovoSeven or FEIBA alone or in combination with Hemlibra using the thrombin generation assay in platelet poor hemophilia A (HA) plasma

Rationale

+ The thrombin generation potential can provide a surrogate marker to assess the potential safety and efficacy of SQ MarzAA in combination with Hemlibra *in vitro* when comparing MarzAA to NovoSeven RT at equipotent concentrations as NovoSeven is generally believed to be safe in combination with Hemlibra

Results

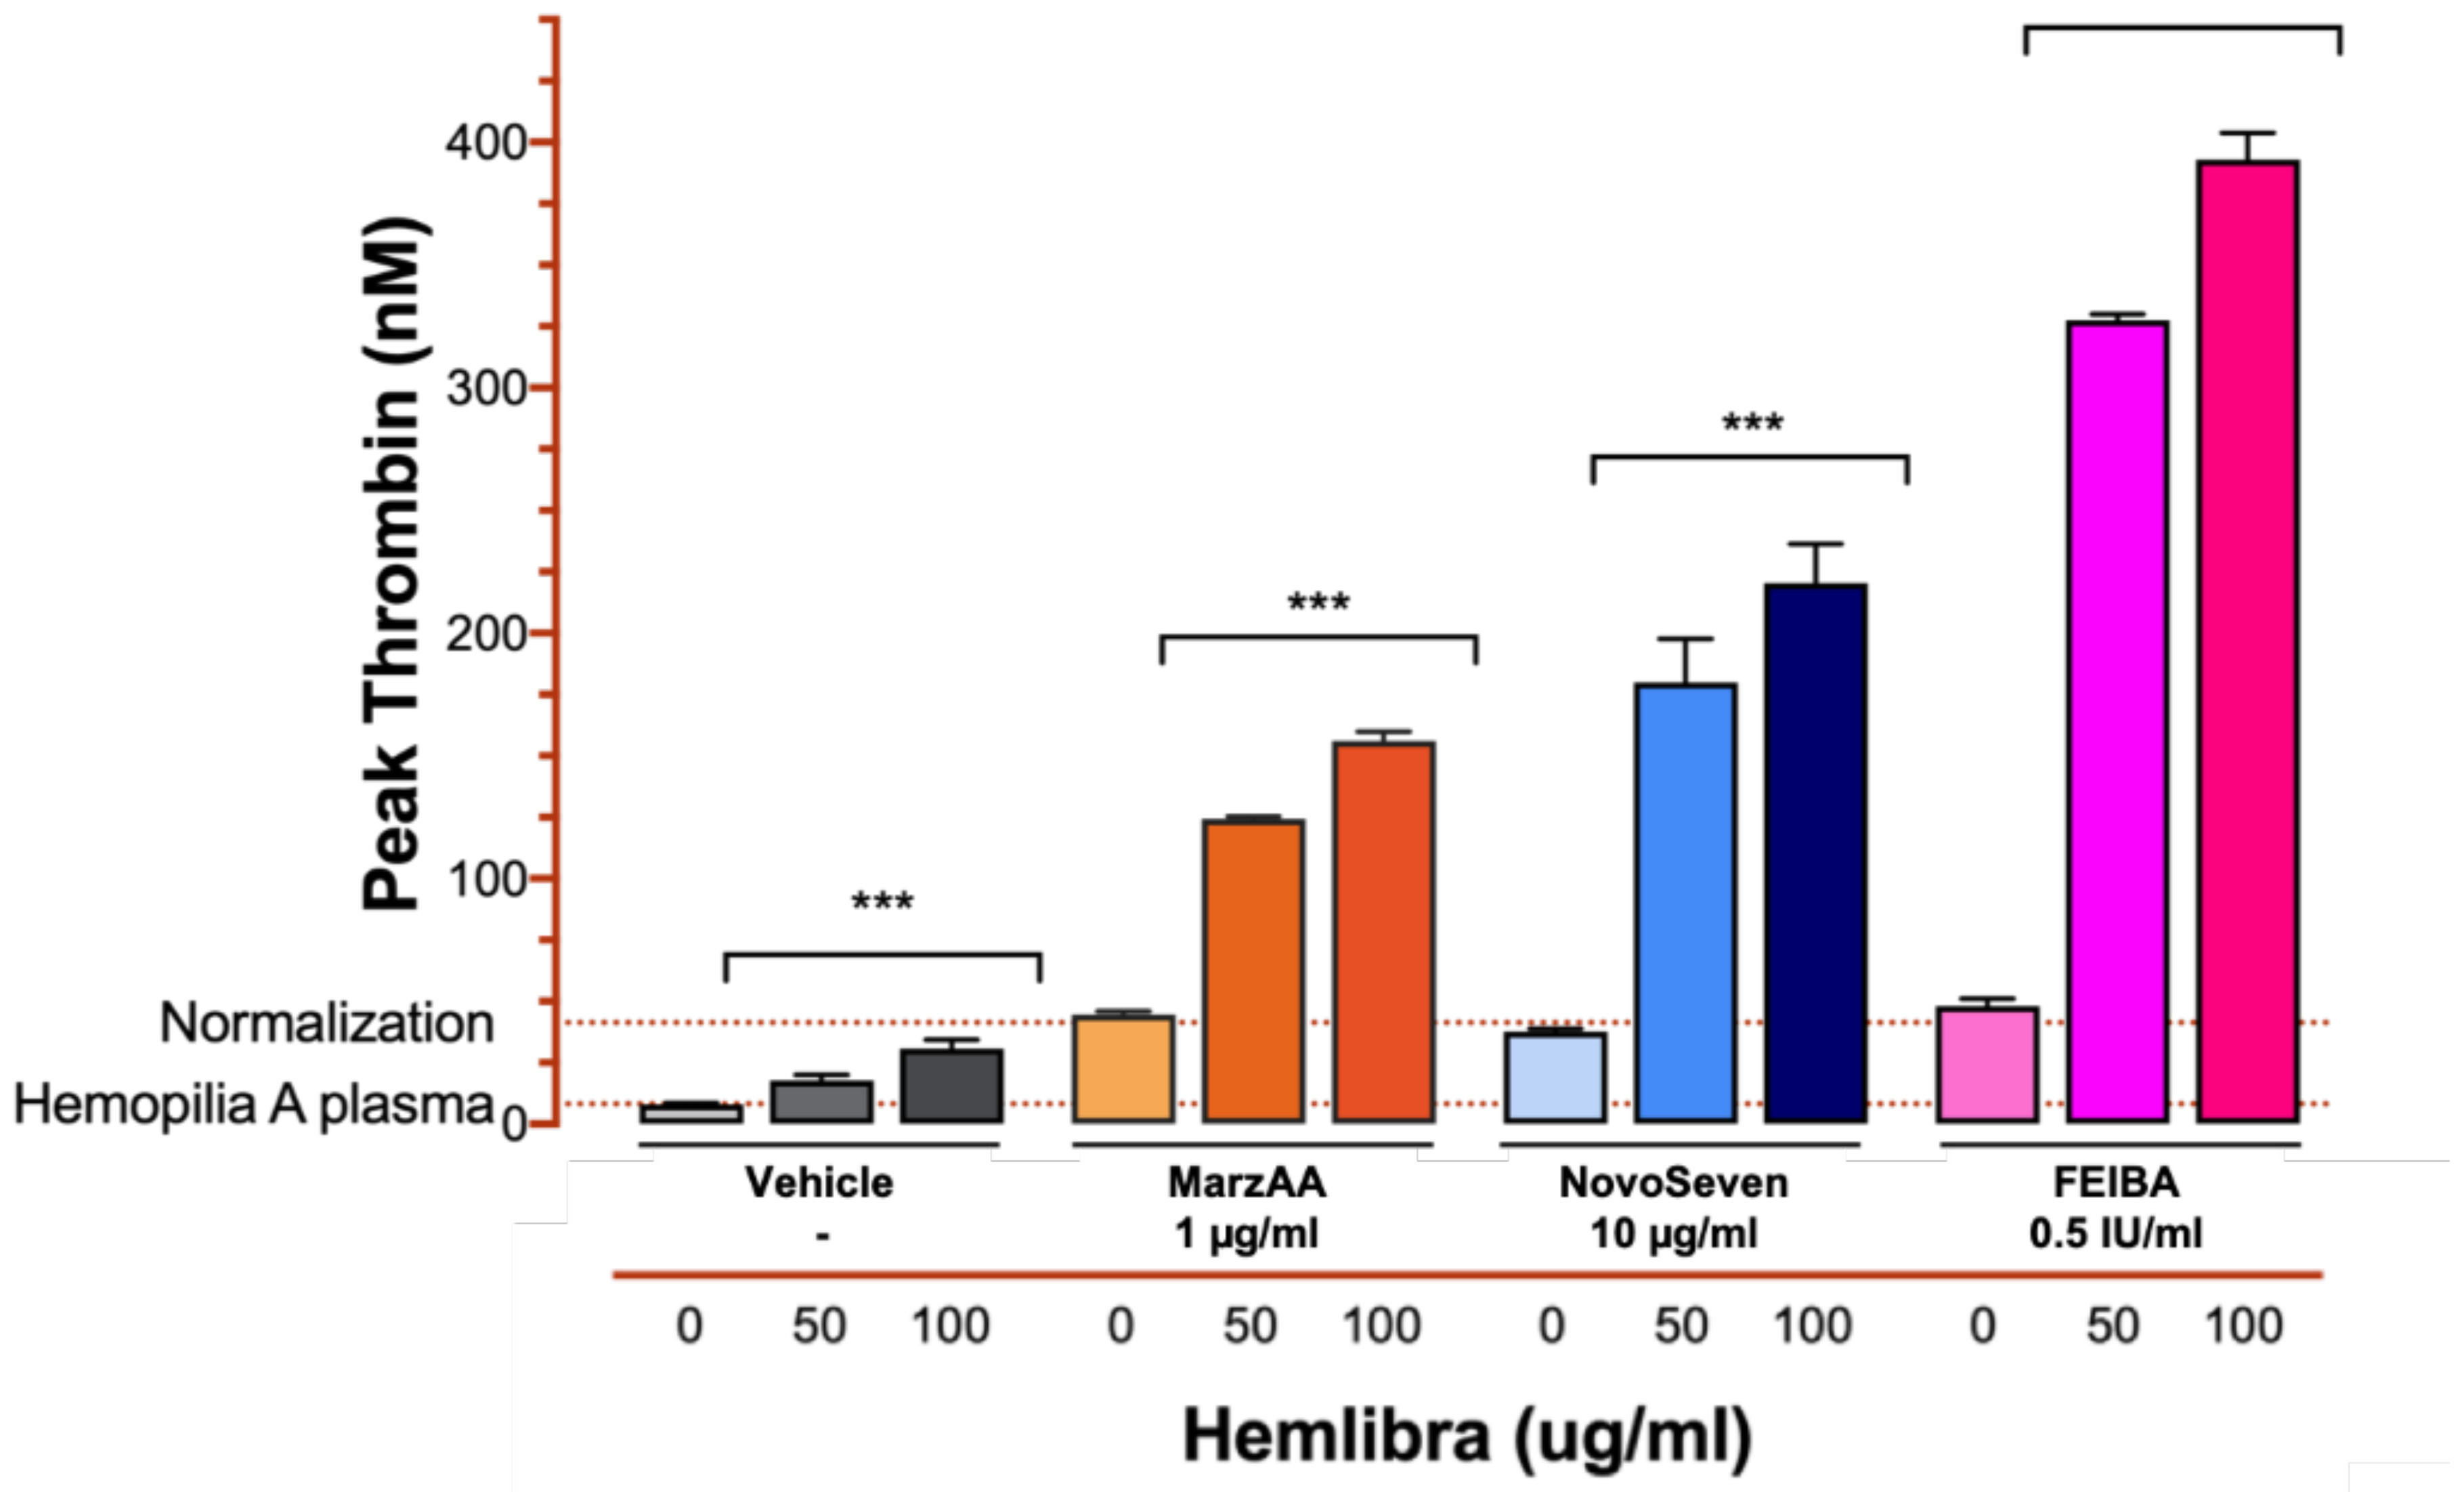
Peak Thrombin (nM)
400
300
200
100
0
Normalization
Hemopilia A plasma

Vehicle
-
MarzAA
1 µg/ml
NovoSeven
10 µg/ml
FEIBA
0.5 IU/ml
0 50 100 0 50 100 0 50 100 0 50 100
Hemlibra (ug/ml)

Fig. 1 Effect of MarzAA, NovoSeven and FEIBA (test compounds) in combination with Hemlibra. Data is shown as mean + SD for buffer (grey), MarzAA (orange), NovoSeven (blue), FEIBA (magenta). Plasma concentrations are indicated below each test compound and the HA plasma is of single donor origin. Hemlibra concentrations for the vehicle and each test compound are shown below the solid maroon line. The dotted lines represent peak thrombin generation levels in the HA plasma (lower line) and in a pooled plasma from healthy individuals (upper line). The asterisks indicate difference from FEIBA at 0.5 IU/mL. It was not possible to go above 0.5 IU/mL FEIBA as it exhausted the assay limits. Data represent duplicate or triplicate experiments run on separate occasions with the same lot of plasma and with each sample run in triplicate within each experiment. Control samples were included on each plate in each experiment.

Key Experimental Messages

+ MarzAA demonstrated an approximate ten-fold increased potency vs NovoSeven
+ Both MarzAA and NovoSeven increase peak thrombin generation in HA plasma to the expected levels observed with normal plasma at the tested concentrations shown in Fig. 1
+ The effect of adding normalizing levels of MarzAA (1 µg/mL), NovoSeven (10 µg/mL) or FEIBA (0.5 IU/mL) to HA plasma containing clinically relevant concentrations of Hemlibra (50-100 µg/mL) was evaluated (Fig. 1)
+ When correcting for the effect of Hemlibra alone, the increase in peak thrombin generation induced by FEIBA was significantly greater than that observed for both MarzAA and NovoSeven (P<0.002)
+ In contrast, the observed increases in thrombin generation for MarzAA and NovoSeven in combination with Hemlibra were statistically indistinguishable
+ FEIBA was not tested at the highest clinically relevant concentration (2.0 IU/mL) as assay limitations were already approached at 0.5 IU/ml, corresponding to ~25% of the plasma concentration expected for a clinical FEIBA dose of 100 IU/kg
+ Concentrations of MarzAA (5 µg/mL) or NovoSeven (50 µg/mL) which for both compounds correspond to ~50 times the expected plasma concentration after standard doses were required before peak thrombin generation became statistically indistinguishable from FEIBA at 0.5 IU/mL when all test articles were evaluated in the presence of Hemlibra (data not shown).

Methods

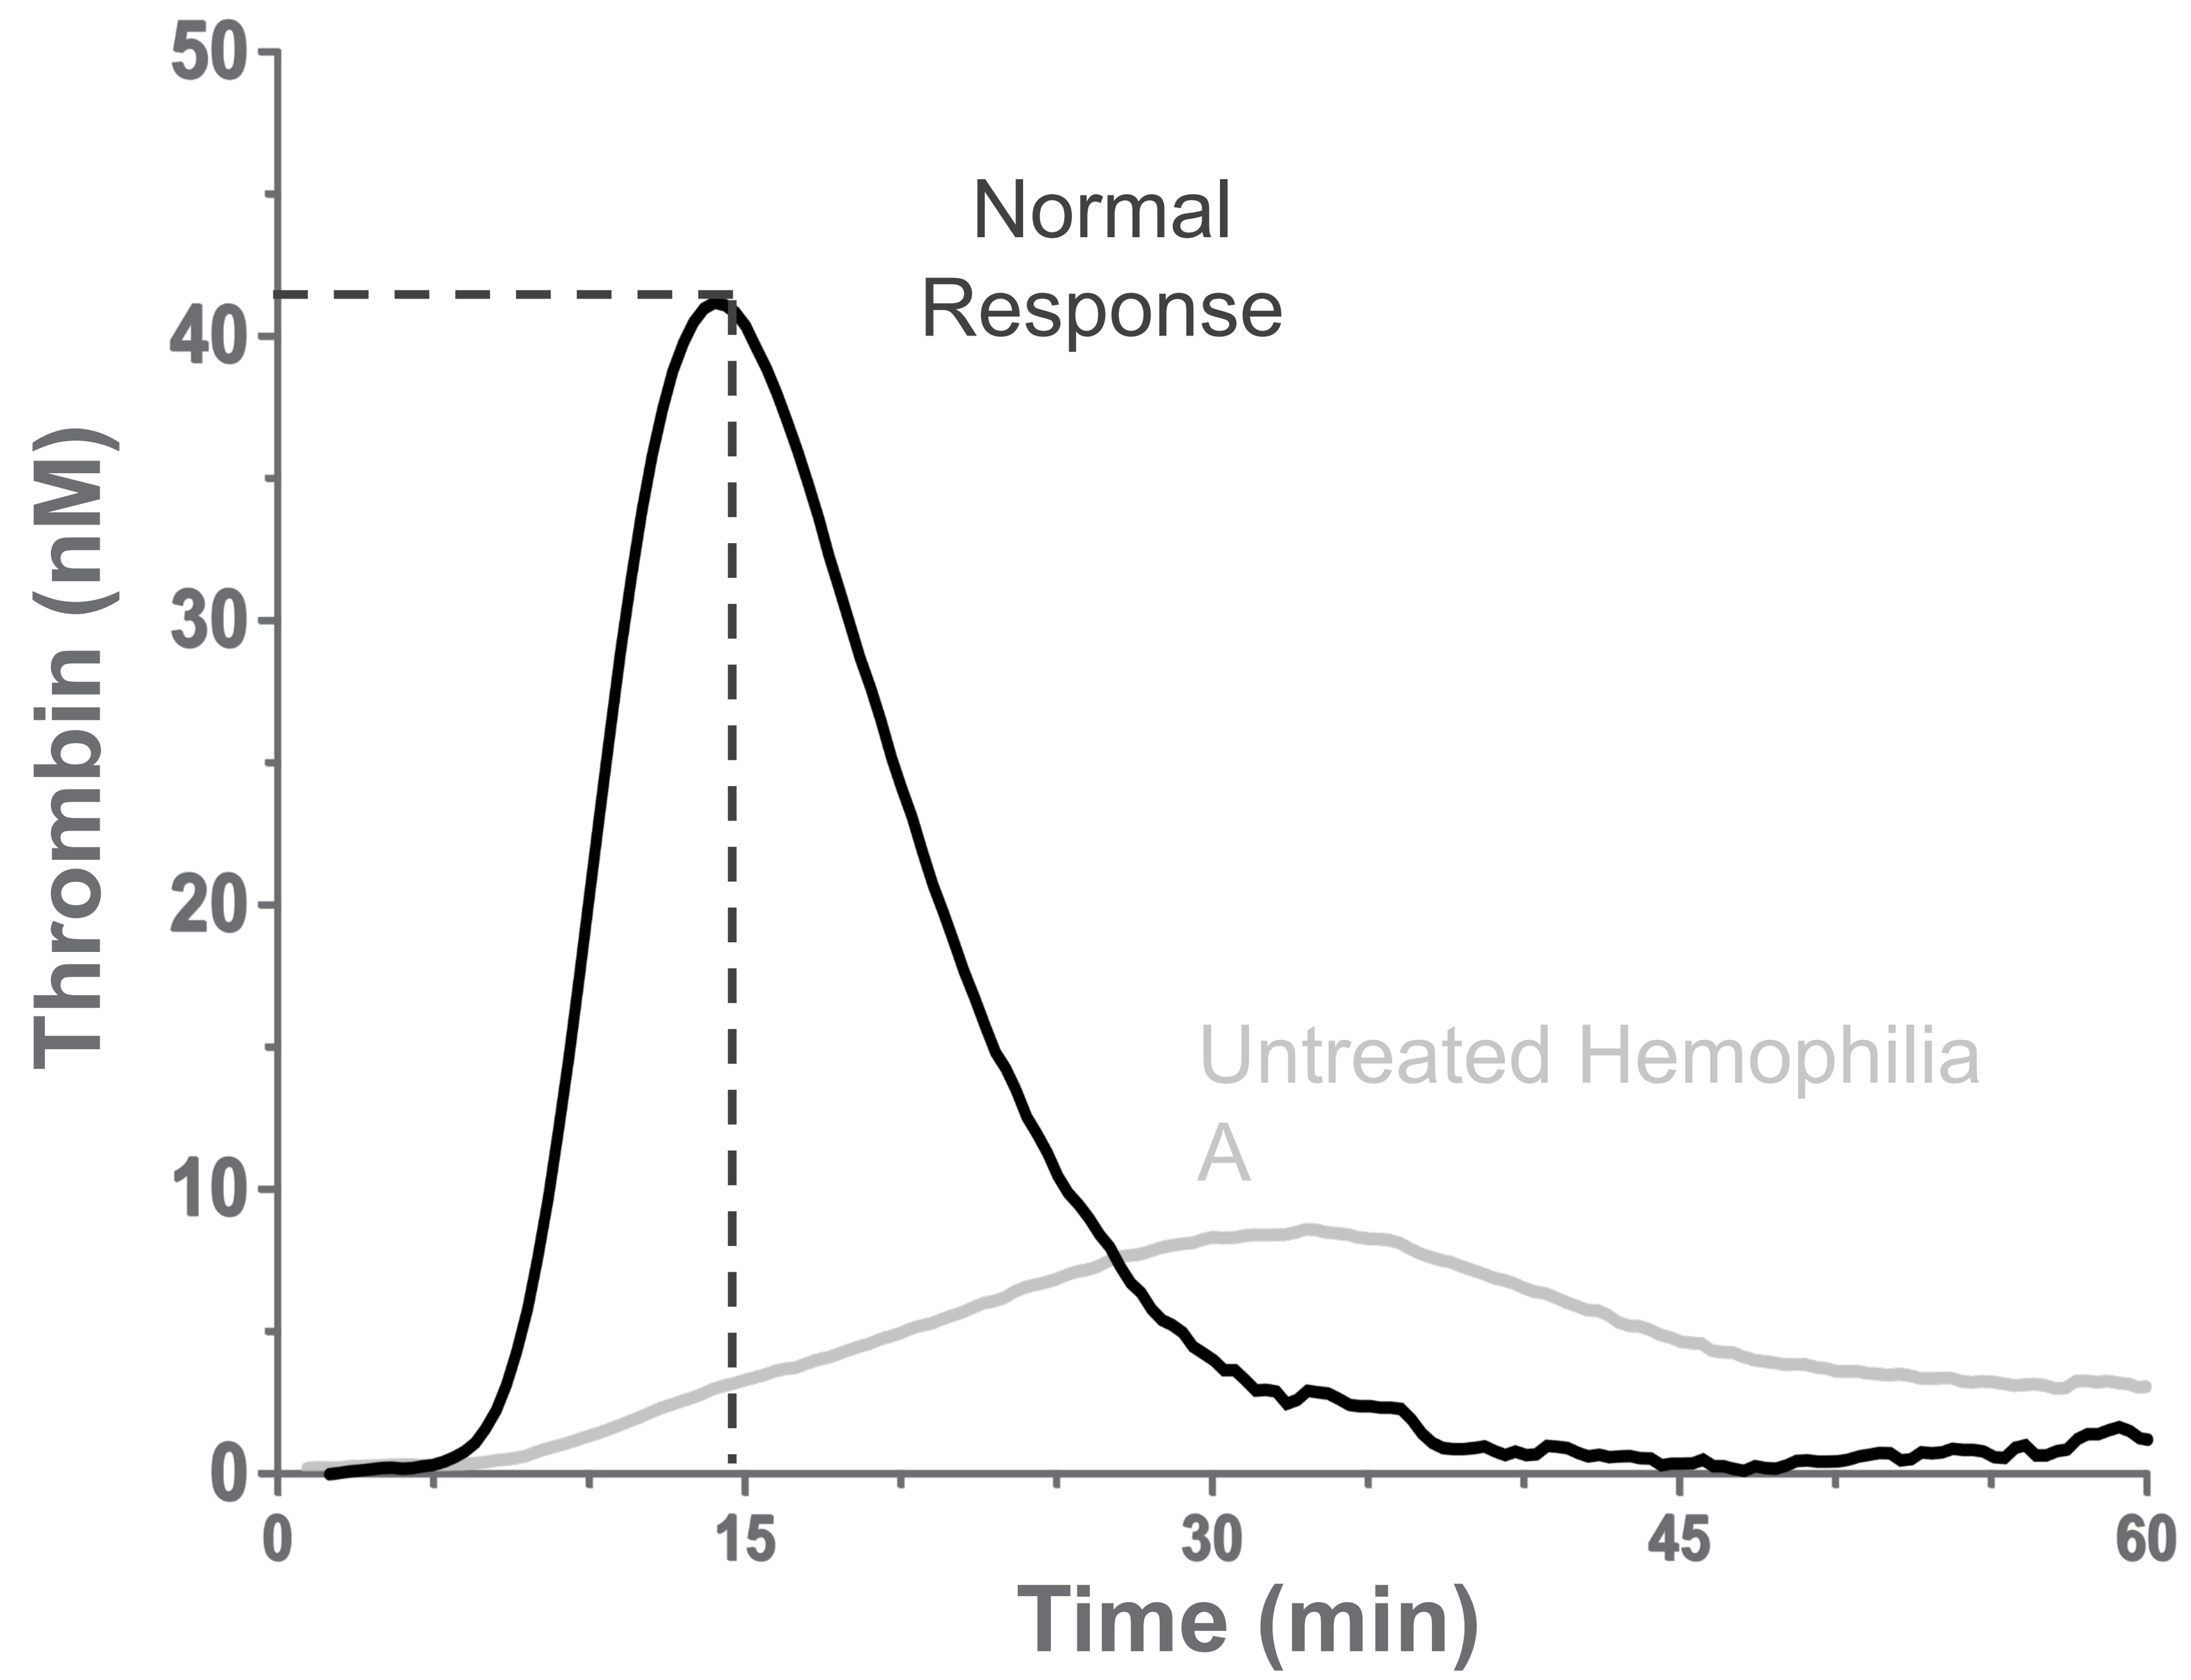
Thrombin (nM)
50
40
30
20
10
0
Normal Response
Untreated Hemophilia A
0
15
30
45
60
Time (min)

Fig. 2 The thrombin generation curve is characterized by an initiation phase (lag-time) followed by the formation of large amounts of thrombin (propagation), culminating in a peak thrombin concentration, and finally inhibition of thrombin generation by natural anticoagulants (Castoldi & Rosing, 2011). Maximal thrombin generation can vary from donor to donor and depending on exact assay conditions; therefore it is important to run comparative experiments in the same matrix.

+ The thrombin generation assay was performed using the standard PPP-Low Tissue Factor and phospholipid containing thrombin generation assay reagent (#TS31.00, Thrombinoscope, Stago)
+ Single donor citrated hemophilia A plasma (George King) was spiked with increasing concentrations of the test articles as described below:
 + FEIBA: 0.25 and 0.50 IU/mL
 + Hemlibra: 0, 25, 50, and 100 µg/mL
 + NovoSeven RT: 0, 1, 2.5, 5, 10, and 50 µg/mL (Eptacog Alfa (Activated))
 + MarzAA: 0, 0.1, 0.5, 1, 2.5, 5, and 10 µg/mL (Marzeptacog Alfa (Activated))
+ Pooled normal control plasma was included to define "normalization" of peak thrombin generation in hemophilia A samples spiked with FEIBA, MarzAA or NovoSeven

Scan to download a copy of the poster

ASH Annual Conference, Abstract #1112 - December 7, 2019